DISCOVERY OF 2-MERCAPTO NICOTINOYL GLYCINE
A NEW POTENT SKIN LIGHTENING AGENT WITH A PROVEN CLINICAL EFFICACY

P. Sextius[1], R. De Dormael[1], G. Lereaux[1], E. Warrick[1], D. Kerob[2], X. Marat[1] and S. Diridollou[1]

[1] L'Oréal Research and Innovation, 1 avenue E.Schueller, 93600 Aulnay sous Bois, France
[2] La Roche-Posay Laboratoire Dermatologique, Levallois-Perret, France

INTRODUCTION

Melanin production inhibitors currently marketed inhibit tyrosinase, the rate limiting enzyme in melanin synthesis process. Yet only few have demonstrated clinical efficacy while some present safety issues. Moreover, environmental respect is barely taken into consideration. Research on skin lightening agents considering efficacy, safety but also environmental respect is then of great importance to significantly improve marketed actives.
For this purpose, a high-throughput screening test (HTS) evaluating melanin production on a large chemical diversity and *in silico* predictive methodology was then used to design original and performant chemical structures resulting in the discovery of 2-mercapto nicotinoyl glycine (2-MNG).
Mechanism studies were conducted *in tubo*, *in vitro*, as well as *in vivo* showing a unique mode of action of 2-MNG consisting in conjugating with melanin precursors, avoiding their integration into growing eumelanin and pheomelanin.

MATERIAL & METHODS

In vitro **screening and 2-MNG discovery:**

- **HTS assay:** Normal human melanocytes (NHM) were cultivated in 384-well plates. Over 100 000 raw materials were screened after dilution in the culture medium at 10 doses and incubation for 72 hours. Optical density was measured at 340 nm reflecting melanin content. Data were normalized in comparison with untreated cells. IC50 were determined.
- **Environmental profile assessment & *in silico* prediction**: Water footprint index (WFI) was determined[1]. An internal digital tool was used to design and screen the best thiopyridinone profiles, taking into consideration physico-chemical parameters, environmental, absorption, distribution, metabolism, excretion and toxicity parameters leading to the selection of 2-MNG.

Mode of action of 2-MNG:

The structure of 2-MNG suggests that it can react with o-quinones to form adducts through the thiol group. Mechanism of action studies were carried to confirm this assumption.

- *In tubo*: Oxidation reactions of melanin precursors by tyrosinase were carried out in presence of 2-MNG. Solutions of L-DOPA, DHICA and DHI were prepared at 100 µM, 100µM and 200µM respectively in 0.05 M sodium phosphate buffer, pH 6.8. 2-MNG was added in each solution at similar concentration. The solutions were mixed with 25 U/ml of mushroom tyrosinase. The oxidation reaction was stopped by adding 1 mL of 1 M HCl (solution 1 and 2), or 200 mg $Na_2S_2O_5$ and 4 ml of 1 M HCl (solution 3). Structures were confirmed by NMR and mass spectrometry.
- *In vitro*: NHM were seeded in 6-well plates and incubated for 72h with increasing concentrations of 2-MNG (7,4 to 200 µM). Supernatants were collected and frozen before analysis. Cell layers were frozen at -80°C. Similar experiment was carried out after adding 700µM of L-Tyrosine in the culture medium. The formation of adducts between 2-MNG and Dopaquinone was monitored in cell lysates and in supernatants by LC/MS analysis.
- **UV-induced pigmentation clinical study**: Formulae containing 2-MNG at 0,5 and 1 % were tested versus their vehicle in a double blinded and randomized clinical study[2]. Briefly, 33 female and male individuals were treated on minizones on the back, five days a week during seven weeks, with 4mg/cm^2 of each formula. During the second week, volunteers were exposed under 0.5MED of UV-daylight during 4 consecutives days. The primary evaluation criteria was colorimetry (Chromameter). The color difference, between verum and placebo was calculated (Delta E).

RESULTS

Selection of 2-MNG

HTS screening led to the discovery of thiopyridinone chemical family as potent brightening structures. 197 analogs were virtually designed and screened based on *in silico* prediction of relevant parameters[3] (safety, environmental respect, skin penetration). 2-MNG (WFI=11) was selected for further investigation as displaying the best compromise between these parameters and WFI compared to on-market benches: 4-n-BR WFI = 5813, kojic acid WFI= 316. (Figure 1)

2-MNG conjugates with DOPA, DHICA, and DHI through the o-Quinones *in tubo*

An oxidation reaction of L-DOPA, DHICA and DHI catalyzed by mushroom tyrosinase in the presence of 2-MNG was carried out. 1H and 13C NMR & MS spectra were performed and revealed the formation of new products, namely 6-MNG-DOPA (92% yield), 4-MNG-DHICA (45% yield) and 2-MNG-DHI, 4-MNG-DHI, 2,4-diMNG-DHI (15, 13 and 5% yield, respectively). This indicated that 2-MNG reacts effectively through the thiol group to o-quinones. (Figure 2)

In vitro confirmation: 2-MNG conjugates with Dopaquinone

The formation of 6-MNG-DOPA was monitored after incubation of melanocytes with increasing concentrations of 2-MNG. 6-MNG-DOPA was detected both in cell lysates and supernatants showing the formation of this adduct in the cell, and its release out of the cell. Addition of L-Tyrosine in the culture medium, increasing melanin production, also increased 6-MNG-DOPA formation and release. (Figure 3)

In vivo efficacy and mode of action of 2-MNG in a UV-induced pigmentation study

The *in vivo* dose effect (0.5 & 1%) of 2-MNG was demonstrated, either to prevent the early darkening or to reduce delayed tanning. This result is connected with 2-MNG capacity to capture melanin precursors; the latter being involved in both pigmentation processes[4]. No adverse effects were reported. (Figure 4)

Figure 1
IN VITRO SCREENING AND VIRTUAL DESIGN LED TO THE DISCIVERY OF 2-MNG

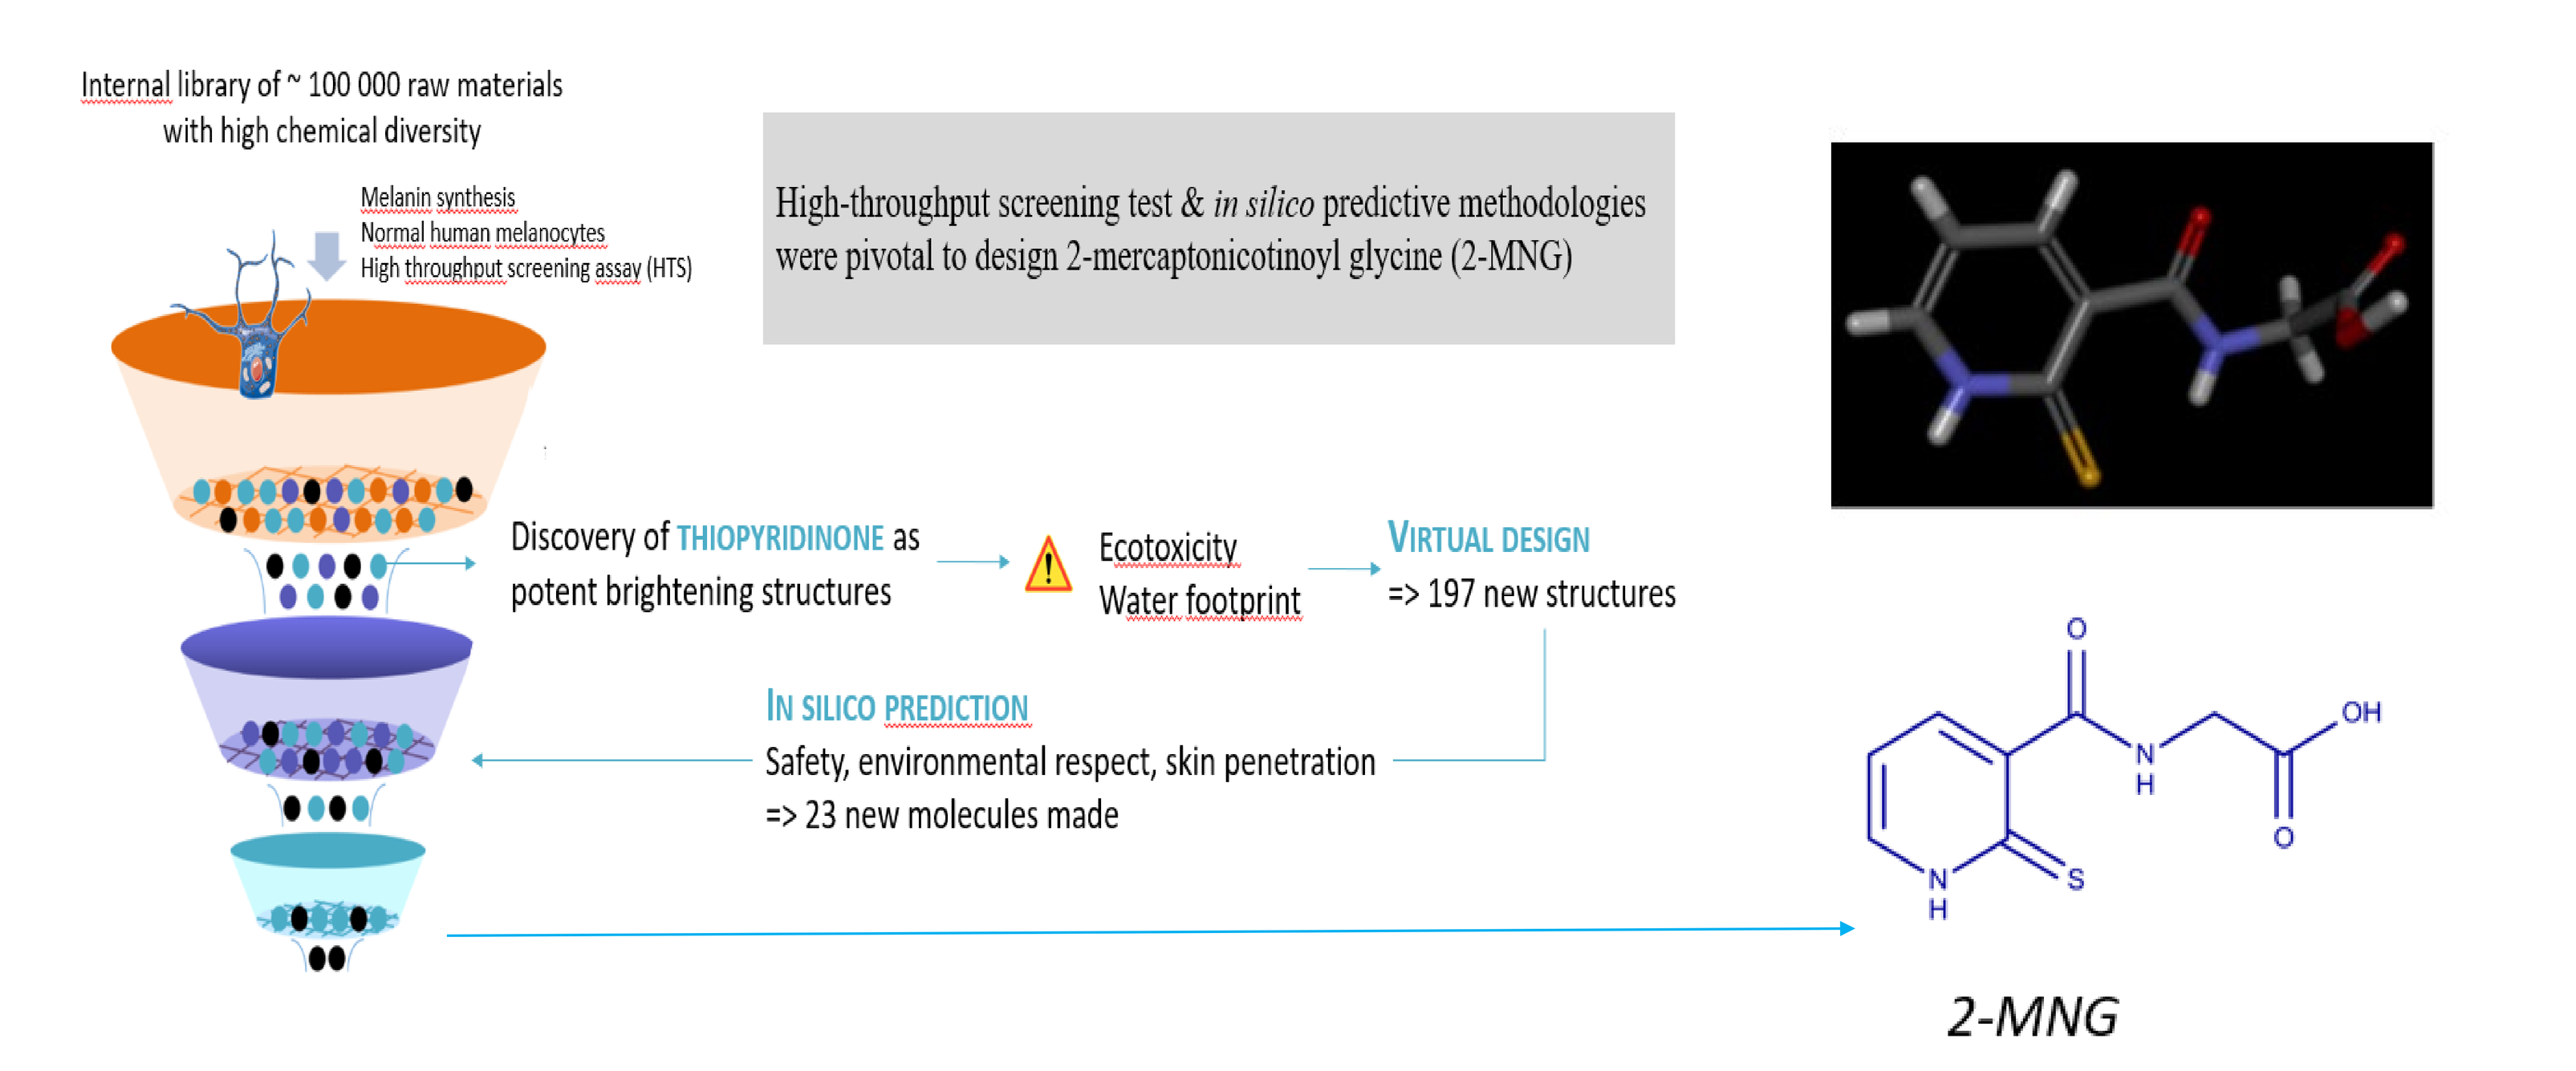
Internal library of ~ 100 000 raw materials
with high chemical diversity
Melanin synthesis
Normal human melanocytes
High throughput screening assay (HTS)
High-throughput screening test & in silico predictive methodologies were pivotal to design 2-mercaptonicotinoyl glycine (2-MNG)
Discovery of THIOPYRIDINONE as potent brightening structures
Ecotoxicity
Water footprint
VIRTUAL DESIGN
=> 197 new structures
IN SILICO PREDICTION
Safety, environmental respect, skin penetration
=> 23 new molecules made
2-MNG

Figure 2
STRUCTURE OF 2-MNG ADDUCTS TO O-QUINONES

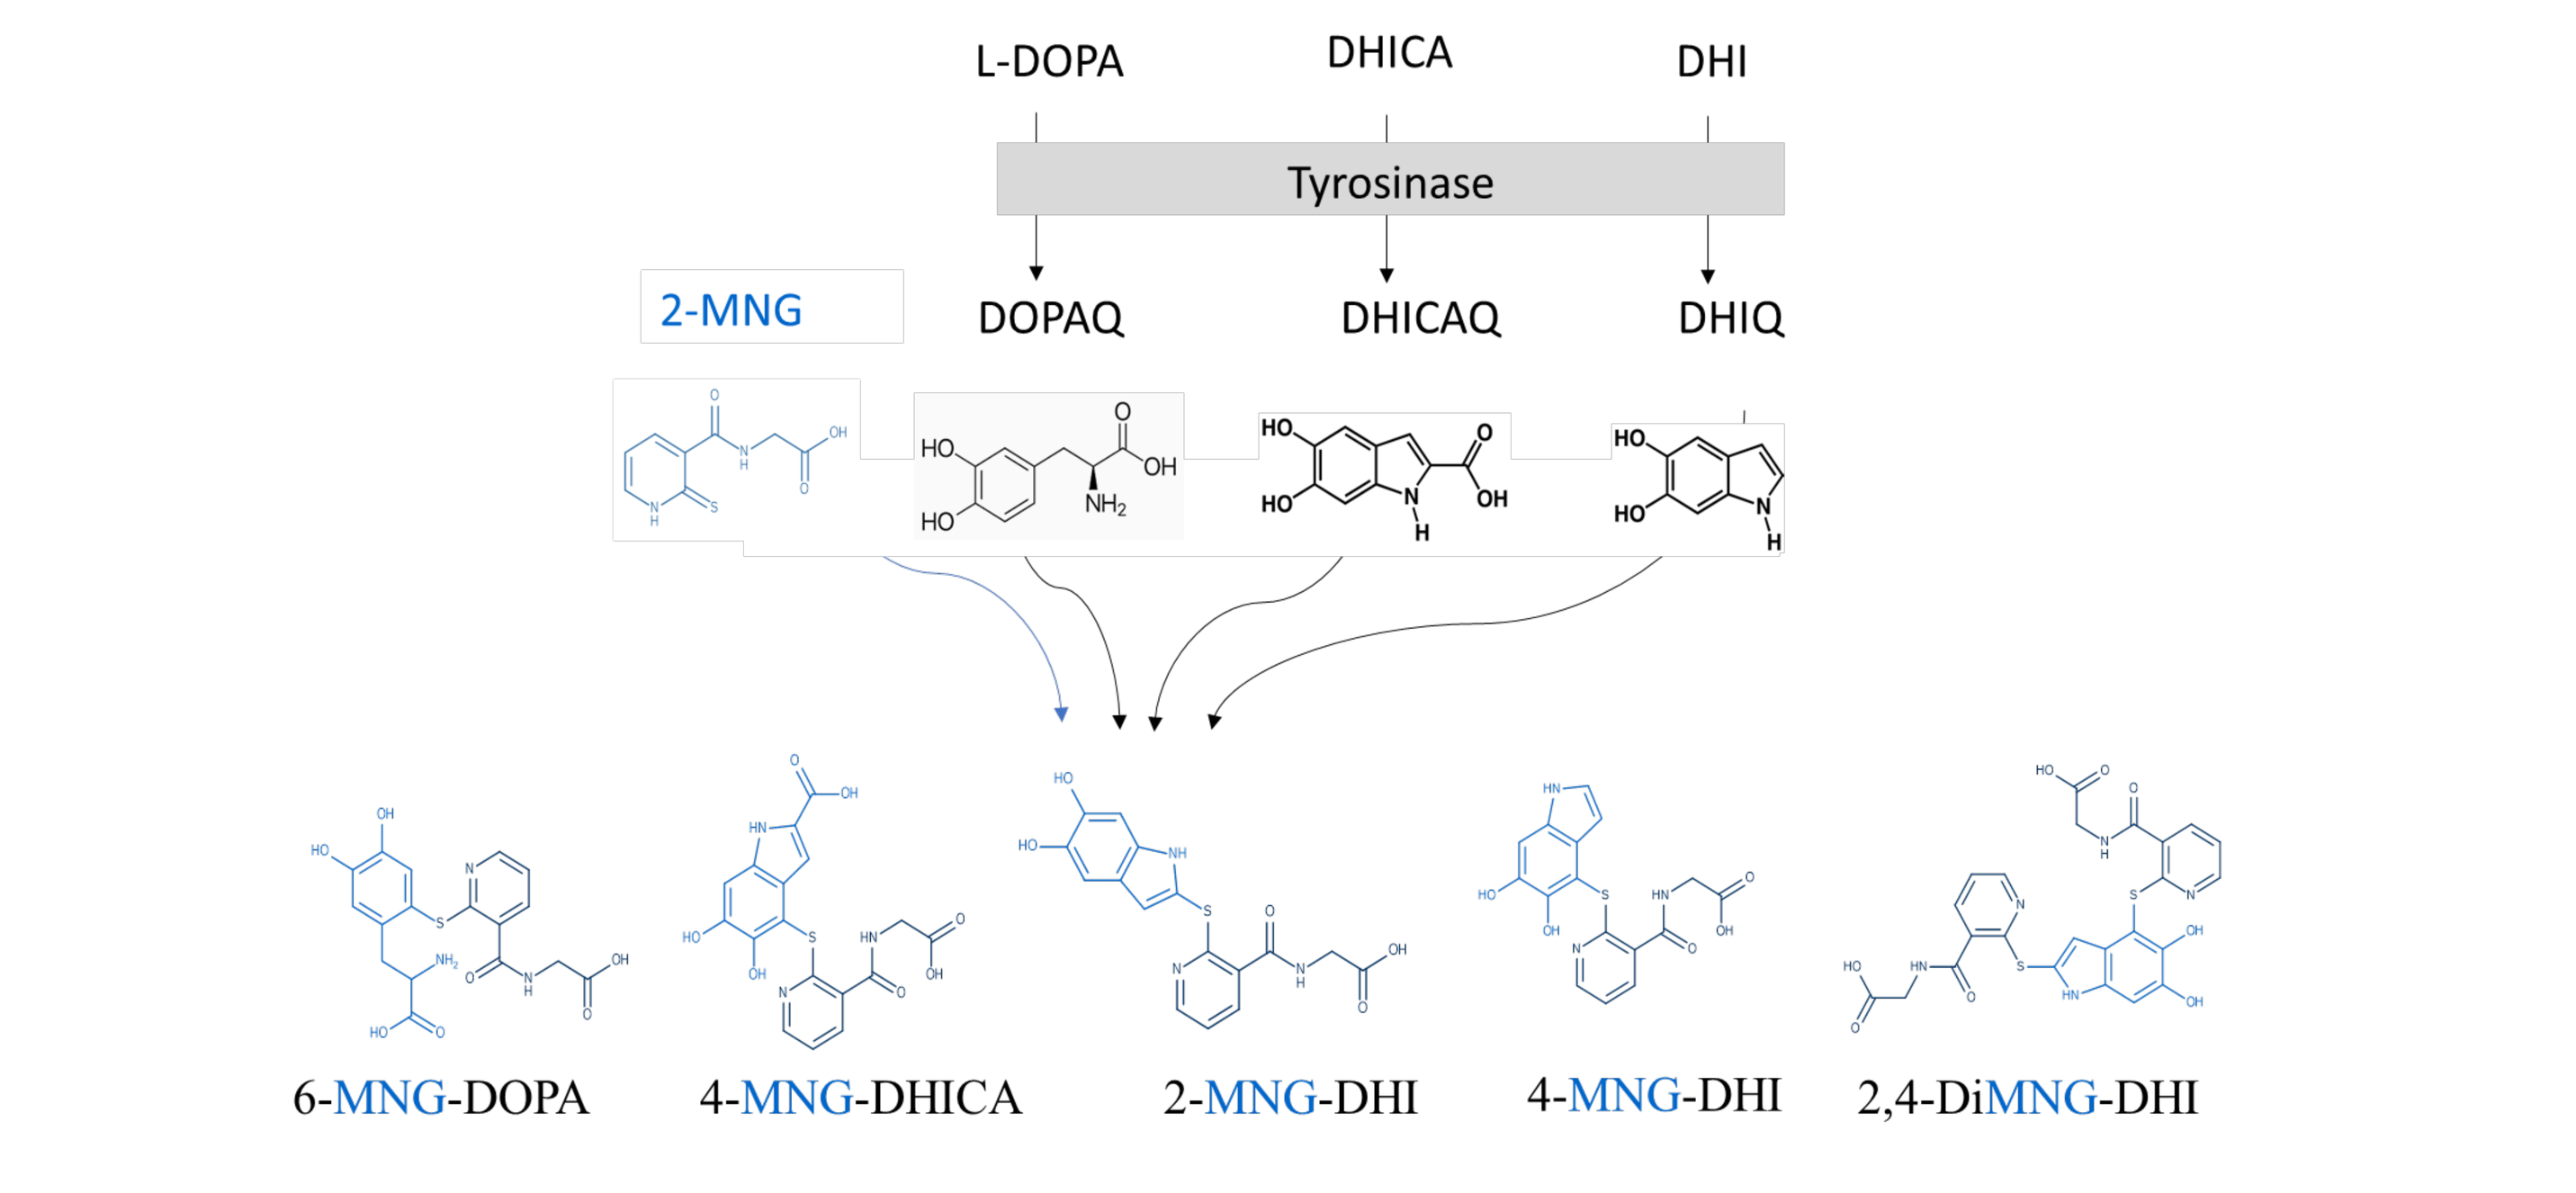
L-DOPA
DHICA
DHI
Tyrosinase
2-MNG
DOPAQ
DHICAQ
DHIQ
6-MNG-DOPA
4-MNG-DHICA
2-MNG-DHI
4-MNG-DHI
2,4-DiMNG-DHI

Figure 3
2-MNG DOSE EFFECT ON MELANOCYTES GROWN WITH (X) OR WITHOUT (△)L-TYR ADDITION.

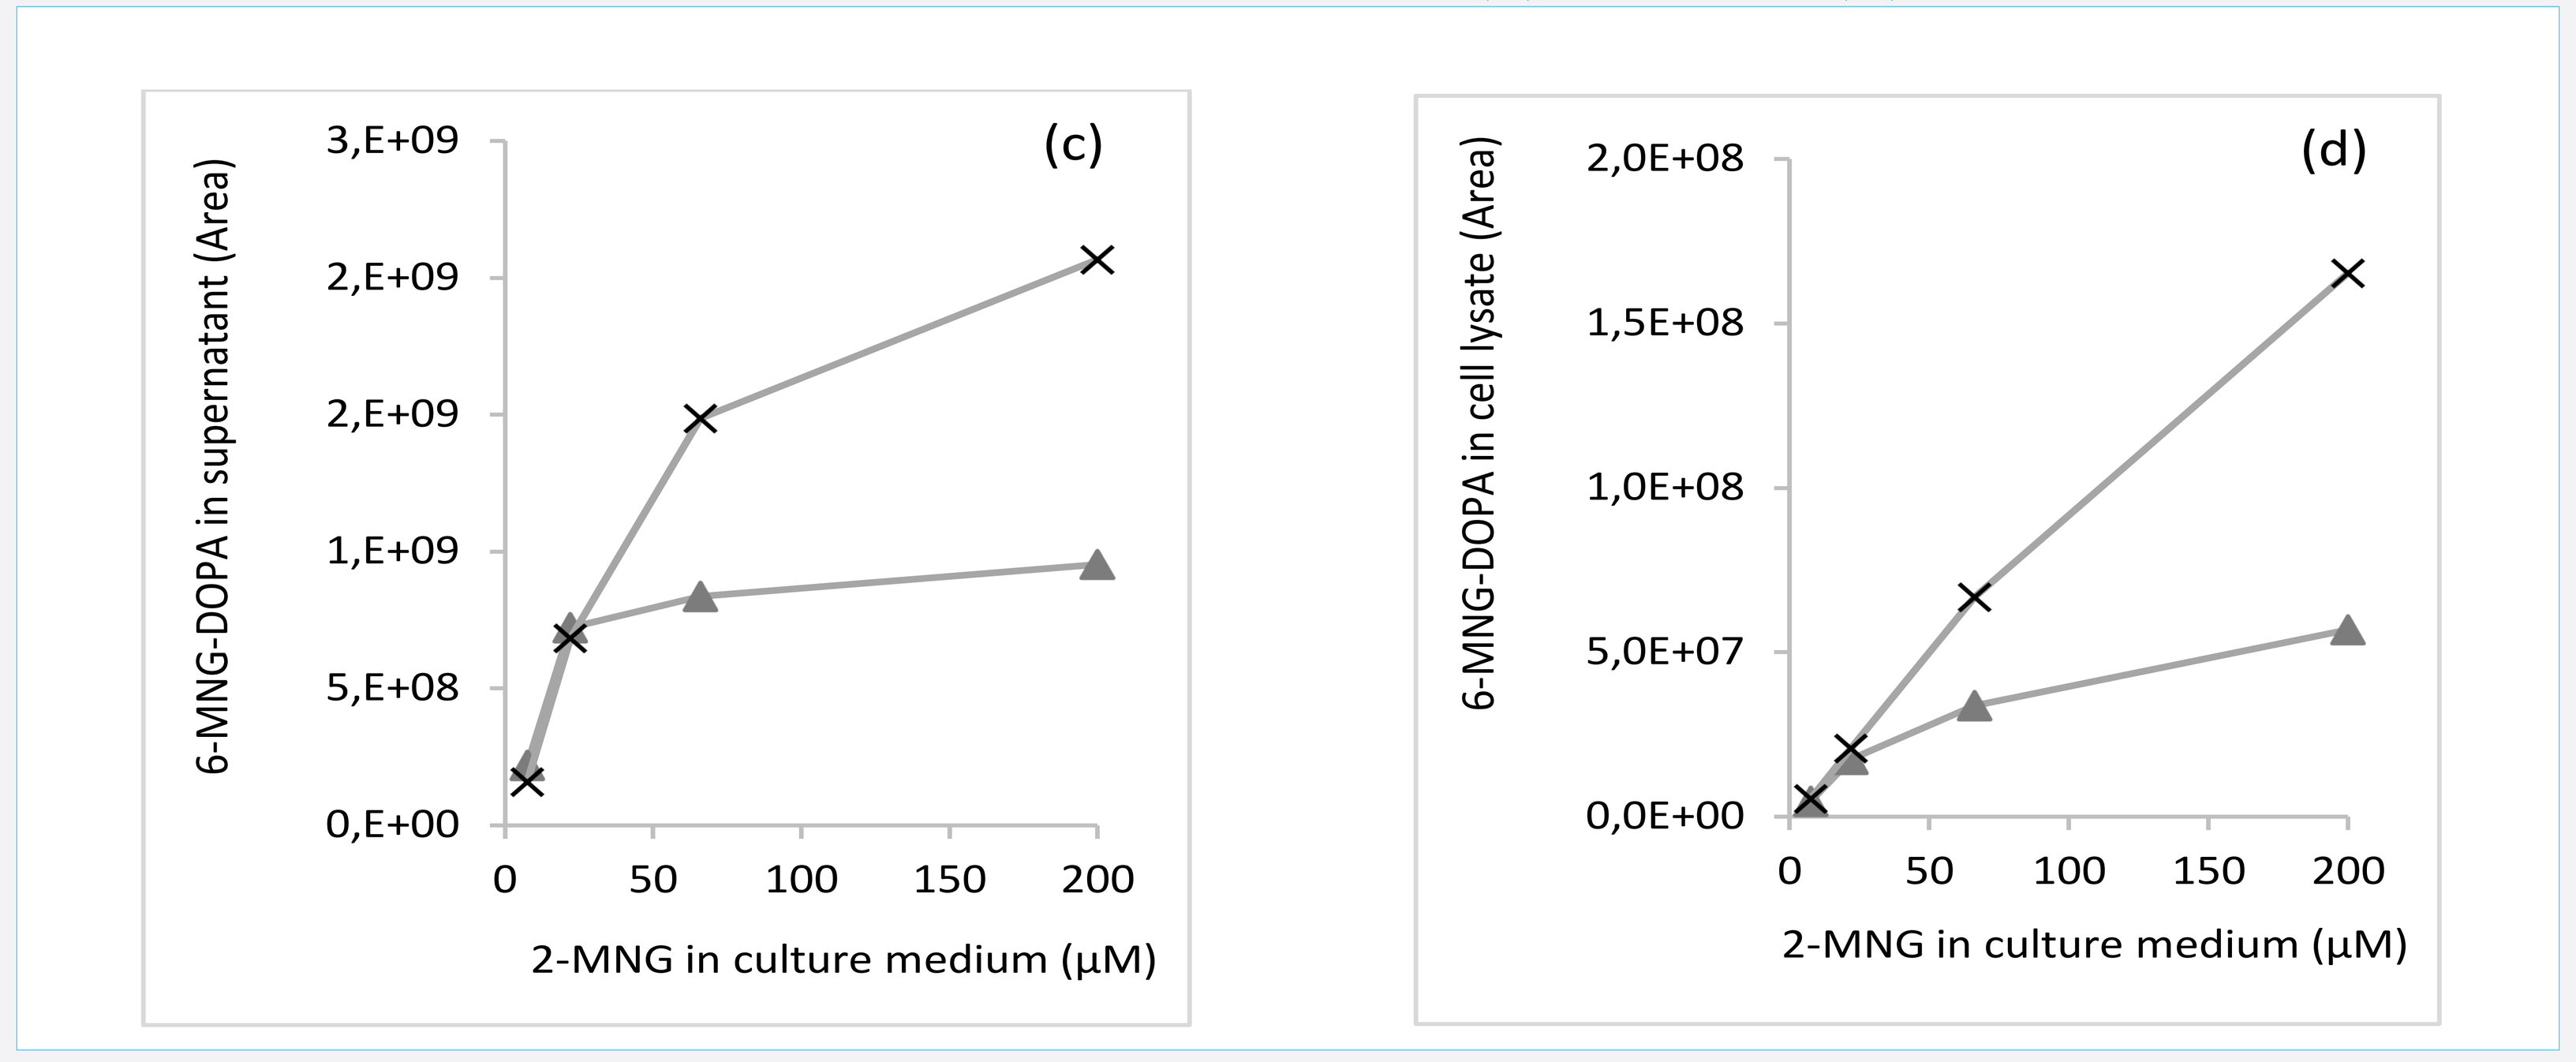
(c)
6-MNG-DOPA in supernatant (Area)
3,E+09
2,E+09
2,E+09
1,E+09
5,E+08
0,E+00
0
50
100
150
200
2-MNG in culture medium (µM)
(d)
6-MNG-DOPA in cell lysate (Area)
2,0E+08
1,5E+08
1,0E+08
5,0E+07
0,0E+00
2-MNG in culture medium (µM)

Figure 4
IN VIVO EFFICACY OF 2-MNG IN A UV-INDUCED PIGMENTATION STUDY

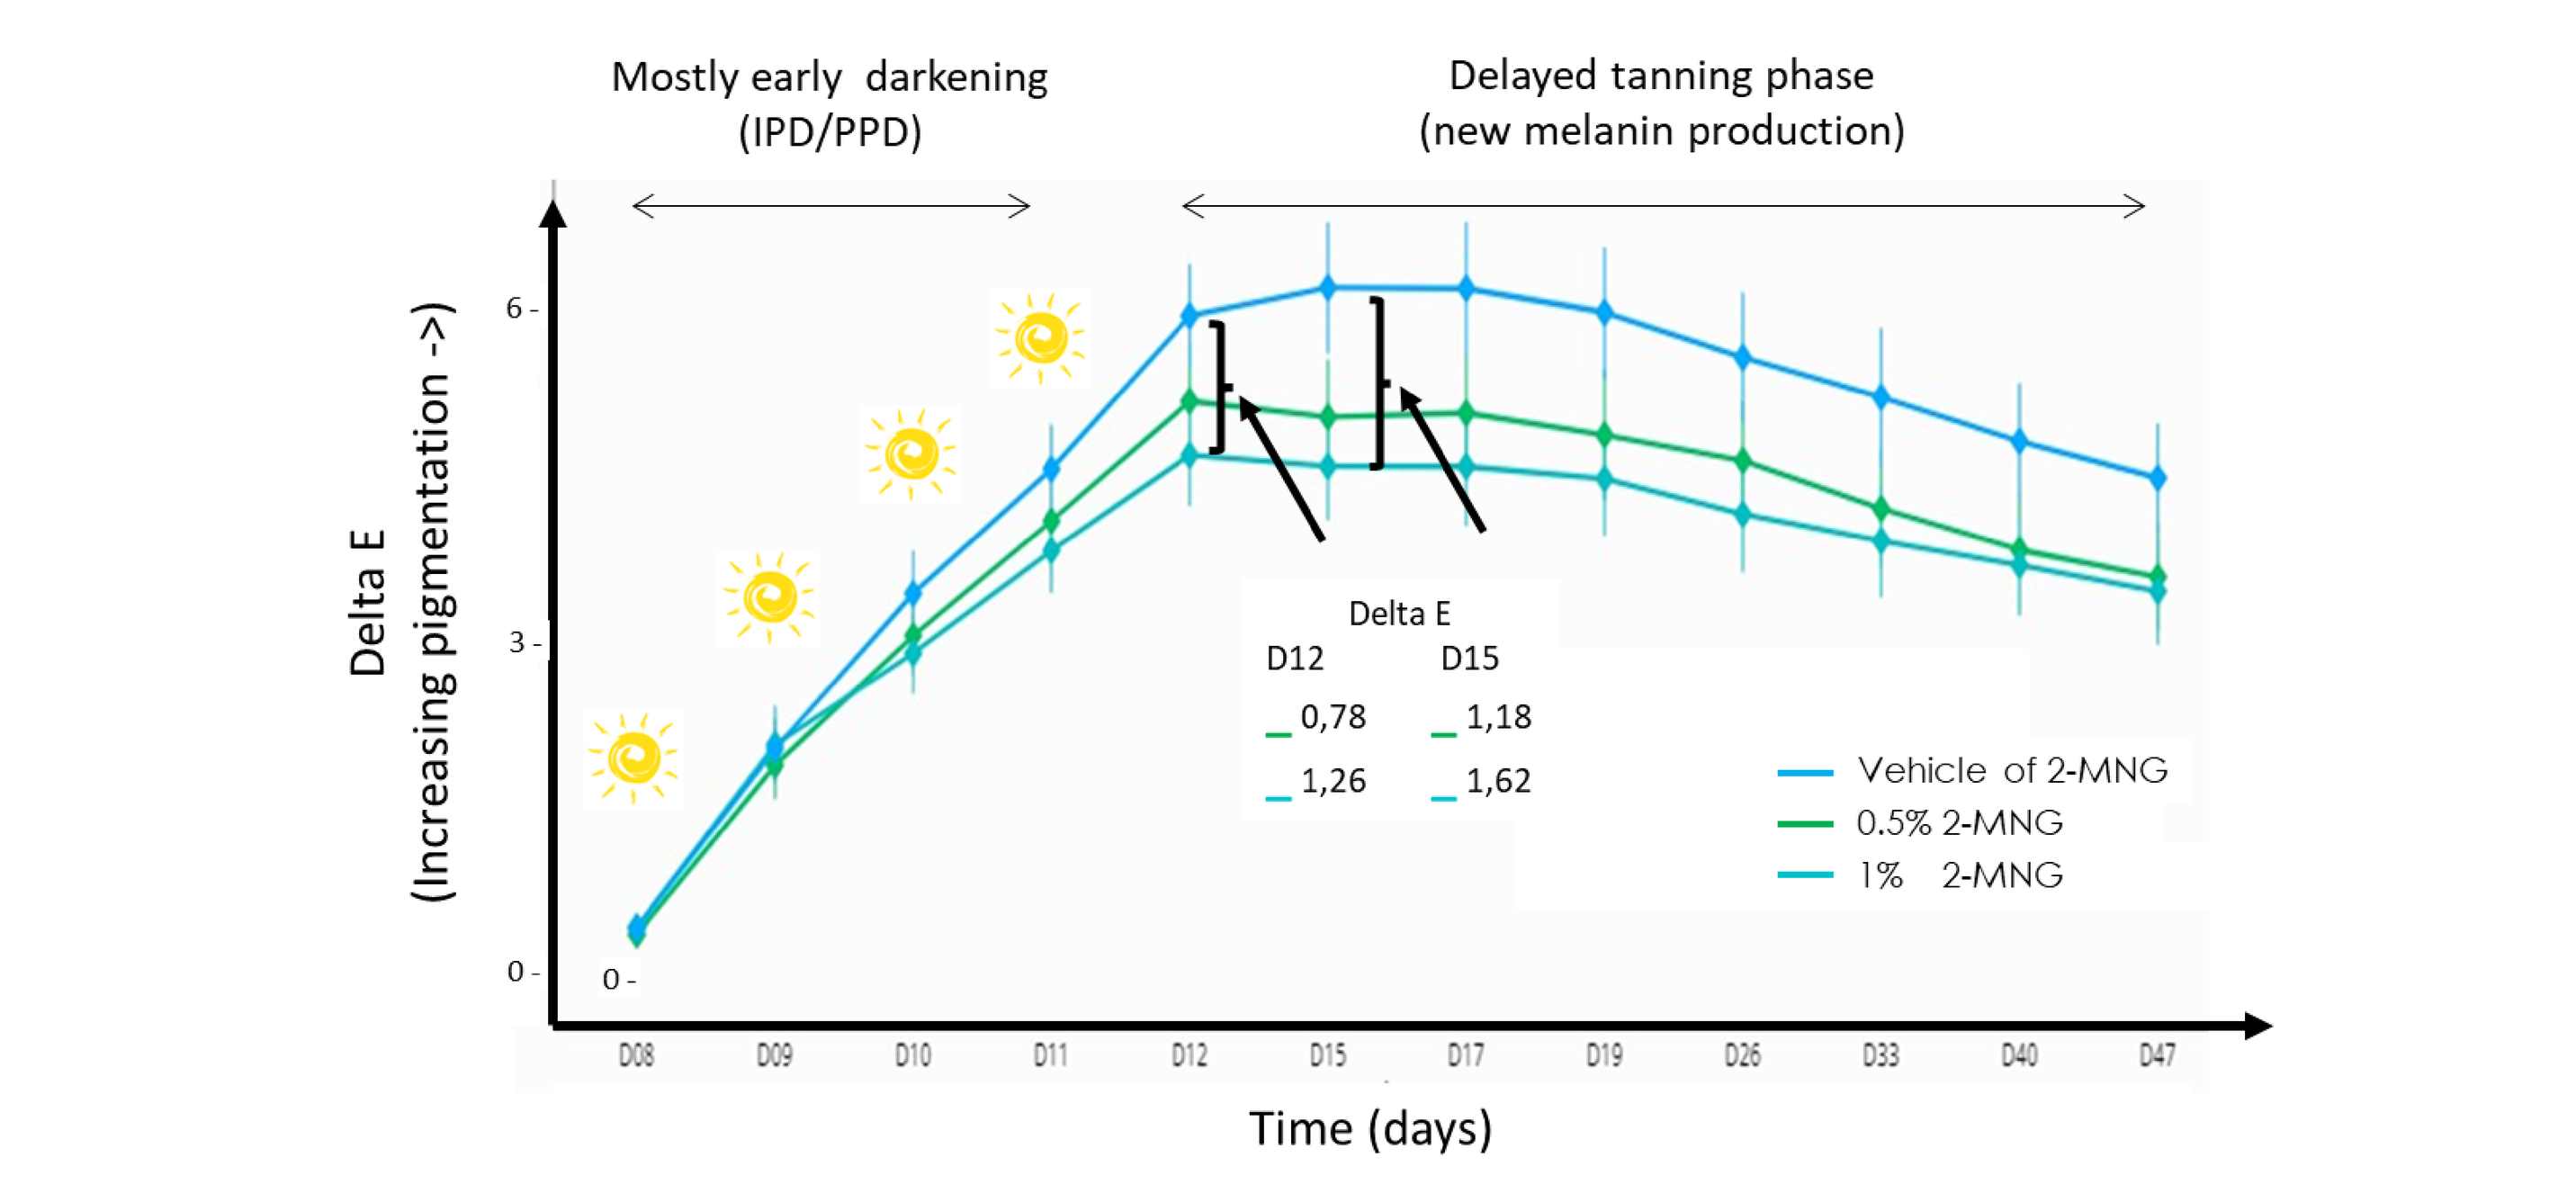
Mostly early darkening
(IPD/PPD)
Delayed tanning phase
(new melanin production)
Delta E
(Increasing pigmentation ->)
Delta E
D12
D15
0,78
1,18
1,26
1,62
Vehicle of 2-MNG
0.5% 2-MNG
1% 2-MNG
D08
D09
D10
D11
D12
D15
D17
D19
D26
D33
D40
D47
Time (days)

CONCLUSION

2-MNG is a new molecule displaying a high efficacy in managing melanin production *in vitro*. Its mode of action, which consists of binding and thus inactivating melanin precursors, leads to a high clinical performance either on prevention of skin darkening or inhibition of delayed tanning with no adverse effects.

References:

1. Ecodesign of cosmetic formulae: methodology and application. International Journal of Cosmetic Science, 40(2), 165-177. https://onlinelibrary.wiley.com/doi/full/10.1111/ics.12448- L'Haridon et al., 2018
2. 2-mercaptonicotinoyl glycine prevents uv-induced skin darkening and delayed tanning. WCD2023 poster - R.deDormael et al., 2023
3. Procédé de dépigmentation des matières kératiniques à l'aide de composés thiopyridinones. Fr 3045604PCMR, Marat, et al., 2015
4. UVA Induced Darkening of Lower Epidermal Cells as an in vitro System of Immediate Pigment Darkening (IPD) and Mechanisms of IPD – Sawamura et al. 1986 https://doi.org/10.1111/j.1346-8138.1986.tb02908.x